Effect of Body Size on the Quantification of Bone Mineral Density from QCT Images Using a Novel Anthropomorphic Hip Phantom

Serena Bonaretti[1], Isra Saeed[1], Andrew J. Burghardt[1], Lifeng Yu[2], Michael Bruesewitz[2], Sundeep Khosla[2], Thomas F. Lang[1]

[1]Musculoskeletal Quantitative Imaging Research Group, Department of Radiology and Biomedical Imaging, University of California, San Francisco, CA, USA
[2]Division of Endocrinology, Metabolism and Nutrition, Department of Internal Medicine, College of Medicine, Mayo Clinic, Rocherster, MN, USA

BACKGROUND

- Pharmaceutical and epidemiological studies often employ quantitative computed tomography (QCT) to estimate volumetric bone mineral density (BMD), bone geometry and strength [1-3].
- Variations in body size and shape, and in bone size and density, result in beam hardening errors that heterogeneously distort image intensities and BMD [4].
- Beam hardening effects may cause BMD measurement artifacts in people of different sizes, and contribute to degradation of data when scanners are changed.

AIM

- To investigate the effect variable body size and bone density on BMD measurements using a novel anthropomorphic hip phantom for different scanners

ANTHROPOMORPHIC HIP PHANTOM (AHP)

- The AHP is designed to study the effect of variable *body size* and *pelvic anatomy* on hip BMD measurements
- It contains a *calibration neck* to correct the measured BMD and a *test neck* to evaluate the correction

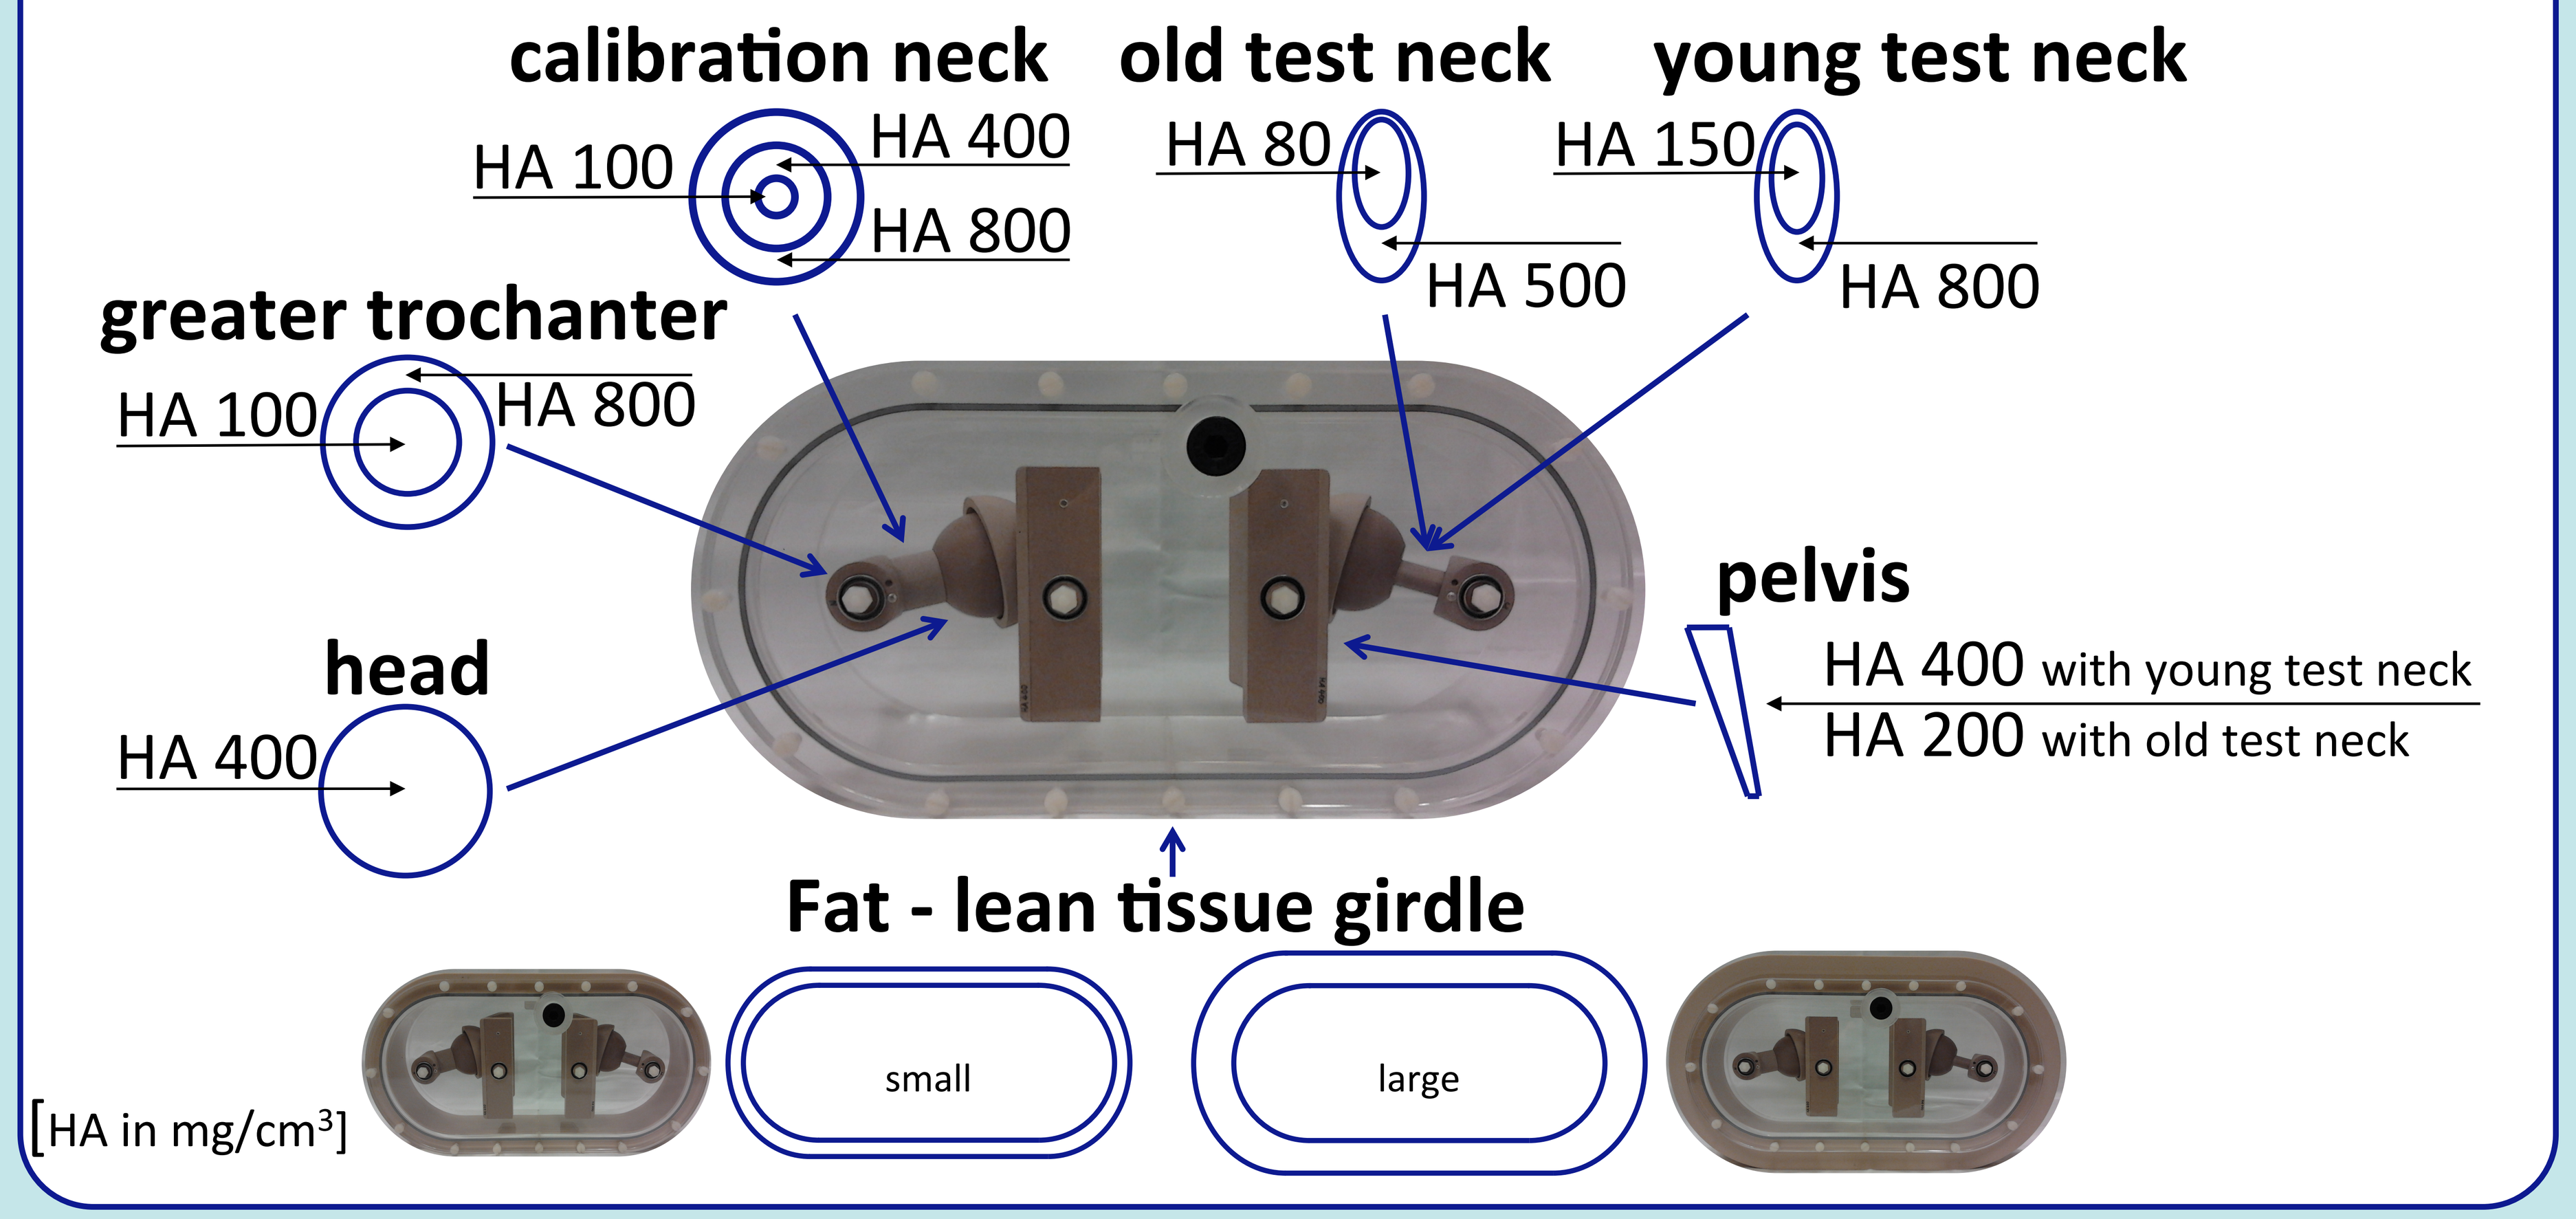
calibration neck
HA 100
HA 400
HA 800
old test neck
HA 80
HA 500
young test neck
HA 150
HA 800
greater trochanter
HA 100
HA 800
head
HA 400
pelvis
HA 400 with young test neck
HA 200 with old test neck
Fat - lean tissue girdle
small
large
[HA in mg/cm³]

EXPERIMENT

- The AHP was scanned in 6 different variations, combining **age** (old/young test hip) and **body size** (no/small/large girdles).
- The scans were acquired on **2 GE VCT 64 systems**, **1 Siemens Biograph** and **1 Siemens Definition,** at UCSF and MAYO Clinic.
- The AHP was scanned with the **patient calibration phantom**, which is commonly used to calibrate subject images in QCT.

MEASUREMENT OF BMD FROM IMAGES

- We calibrated each image with the calibration phantom
- We calculated cortical, trabecular and integral BMD for calibration and test hips
- We chose the Siemens Biograph as reference for its less dependence to body size
- We computed regression equations of calibration hip BMD for each scanner against reference scanner
- We applied calibration hip regression to each image and compared pre- and post correction measurements

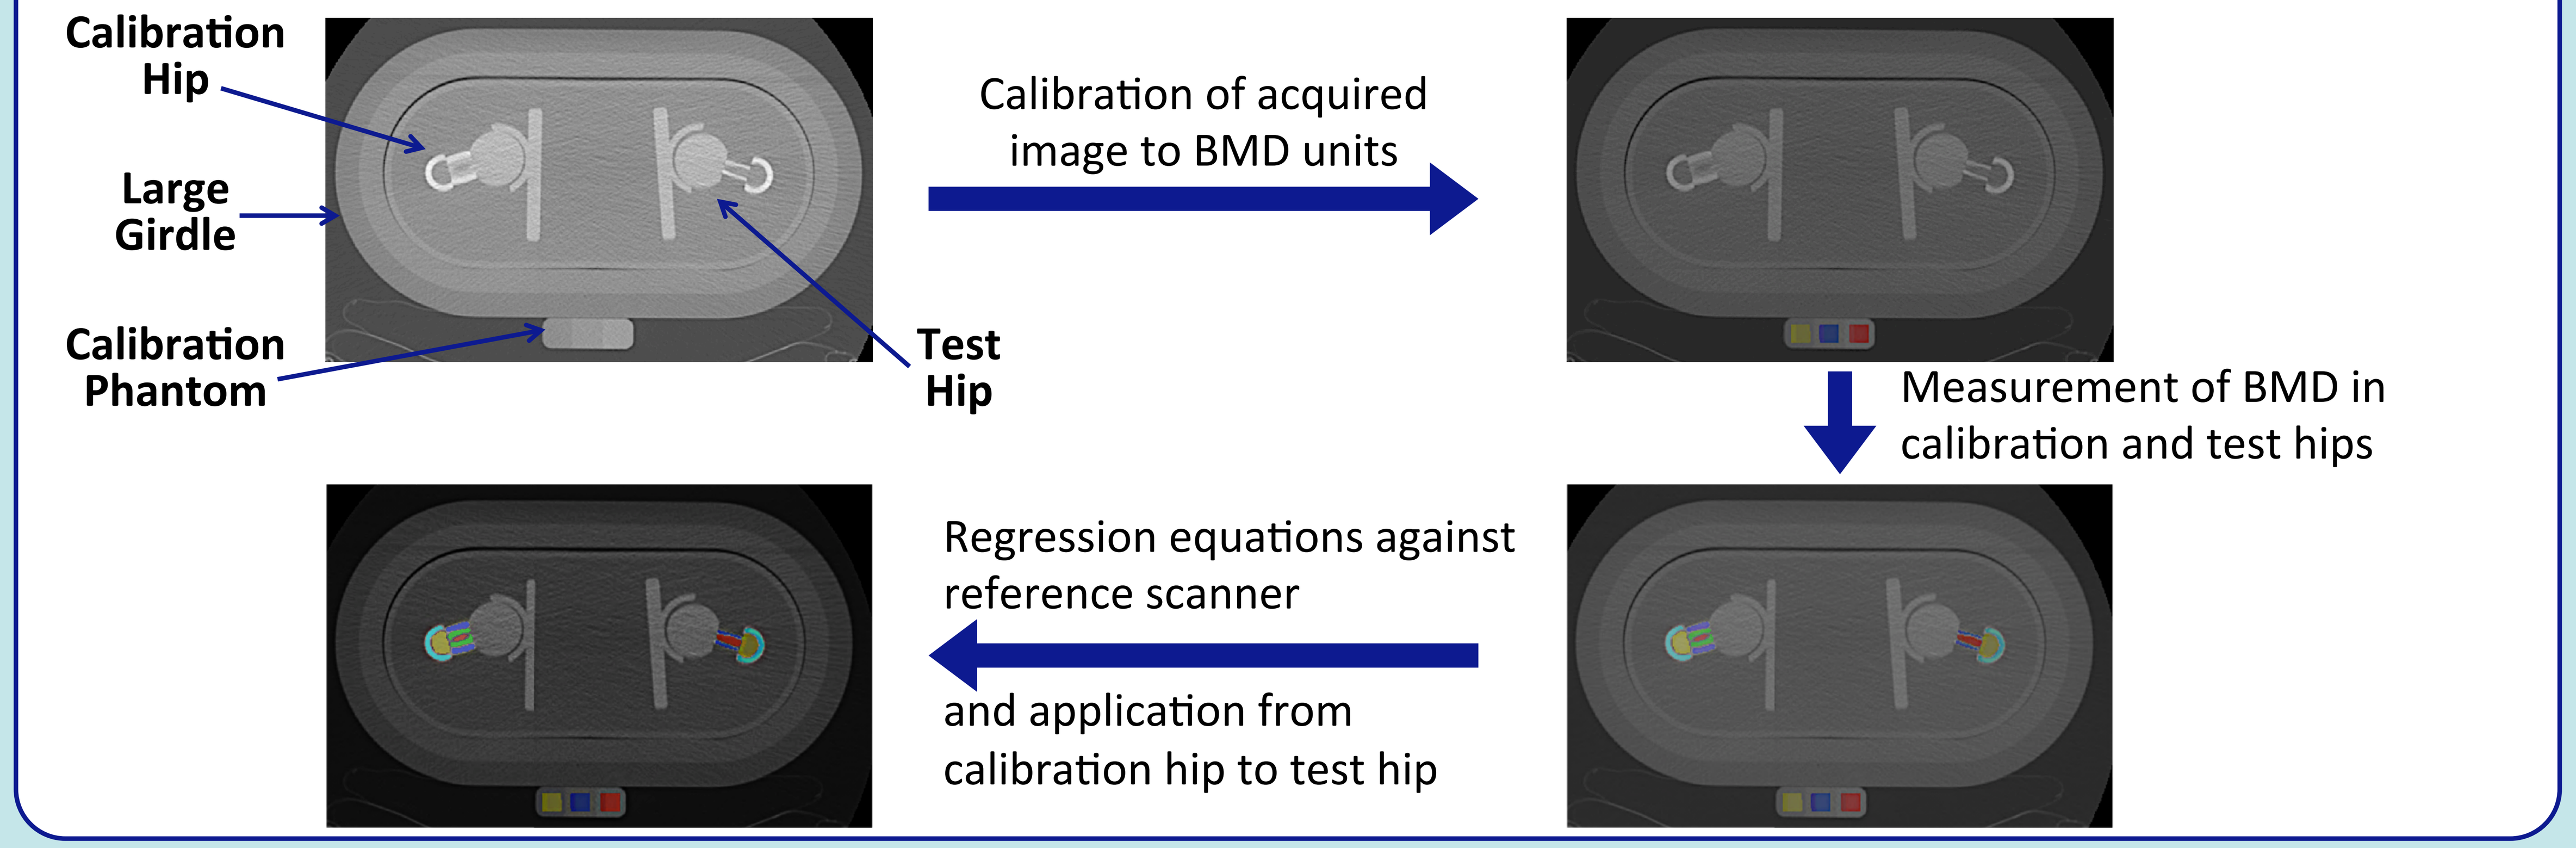
Calibration Hip
Large Girdle
Calibration Phantom
Test Hip
Calibration of acquired image to BMD units
Measurement of BMD in calibration and test hips
Regression equations against reference scanner
and application from calibration hip to test hip

BMD OF TEST HIP BEFORE AND AFTER CORRECTION

- Larger BMD differences were found of the **cortical bone** of femoral neck and greater trochanter

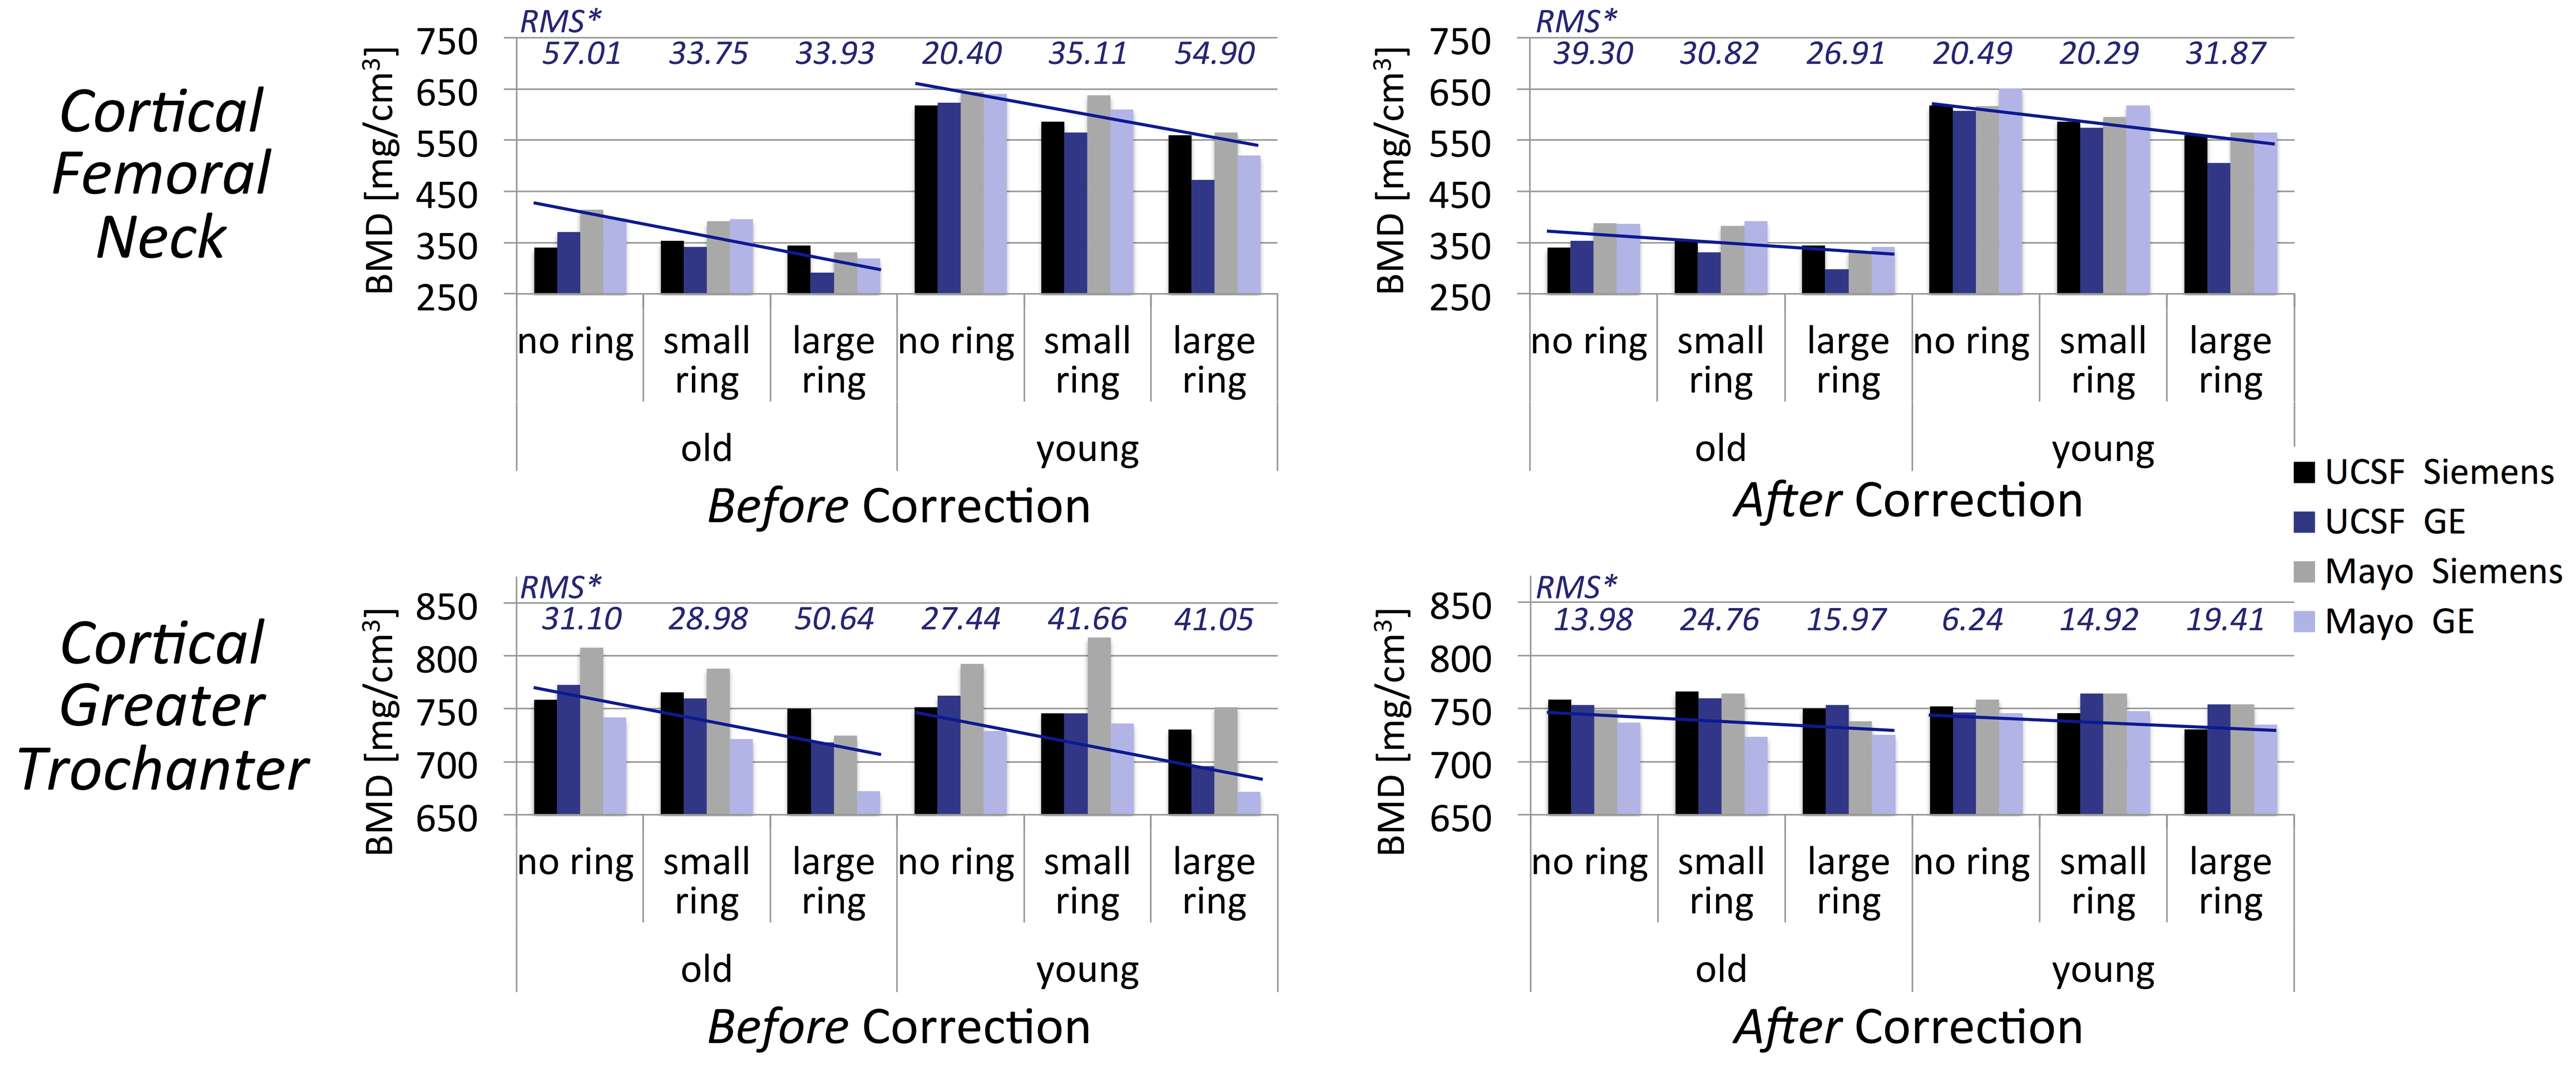
Cortical Femoral Neck
BMD [mg/cm³]
RMS*
57.01 33.75 33.93 20.40 35.11 54.90
no ring
small ring
large ring
old
young
Before Correction
39.30 30.82 26.91 20.49 20.29 31.87
After Correction
Cortical Greater Trochanter
31.10 28.98 50.64 27.44 41.66 41.05
13.98 24.76 15.97 6.24 14.92 19.41
UCSF Siemens
UCSF GE
Mayo Siemens
Mayo GE

*RMS** = root mean square of inter-scanner differences. For each ring configuration, inter-scanner differences were calculated between the reference scanner and the other scanners.
—— = Trend of BMD with body size

DISCUSSION

In this study we analyzed the consequences of *variable body size* on *BMD measurements* using the AHP. We found that:

- Values of cortical BMD decrease dramatically with increasing body size
- Inter-scanner differences are body size dependent, especially for cortical BMD
- Body size dependence was more visible for the GE VCT 64 systems and the Siemens Definition
- Corrections based on calibration hip dramatically reduced inter-scanner errors

Implications:

- Artifactual reductions of BMD with increasing weight complicate measurements in obese patients and patients undergoing weight loss
- Special consideration is required for bone strength estimates. Elastic modulus estimates vary with ≈BMD^2, and cortical voxels are located farthest from neutral axis
- Results show poorer efficacy for correcting thin structures such as cortical femoral neck. We are carrying out studies to address potential interaction between spatial resolution and inter-scanner/body weight variations.
- Overall, initial results show promise for phantom-based corrections to reduce variations in BMD between scanners and patients

APPLICATION TO HUMAN SUBJECTS

- We are currently working on applying the inter-scanner corrections to human subjects, where the same subject was scanned on two different scanners
- Analyses of body size and inter-scanner corrections on bone strength are currently underway.

REFERENCES

1. Black D.M. et al. *Proximal femoral structure and the prediction of hip fracture in men: a large prospective study using QCT*. J Bone Miner Res. 2008. 23(8):1326-33
2. Keaveny T. et al. *Femoral and vertebral strength improvements in postmenopausal women with osteoporosis treated with denosumab*. J Bone Miner Res. 2013 Jun 21
3. Eastell R. et al. *Effect of once-yearly zoledronic acid on the spine and hip as measured by quantitative computed tomography: results of the HORIZON Pivotal Fracture Trial*. Osteoporos Int. 2010. 21(7):1277-85
4. Hsieh, Jiang. *Computed tomography*. Bellingham, Wash. SPIE, c2009

MORE INFORMATION

Information and technical details about this study are available at https://sites.google.com/site/serenabonaretti/asbmr2013

Acknowledgment

The project is funded by NIH/NIAMS 5R01AR060700

serena.bonaretti@ucsf.edu