Comparability of HR-pQCT Bone Quality Measures Improved by Scanning Anatomically Standardized Regions

Serena Bonaretti[1], Margaret Holets[2], Isra Saeed[1], Louise K. McCready[2], Thomas M. Lang[1], Sundeep Khosla[2], Andrew J. Burghardt[1]

[1]MQIR, Department of Radiology & Biomedical Imaging, University of California San Francisco CA, USA
[2]Division of Endocrinology, Metabolism and Nutrition, Department of Internal Medicine, College of Medicine, Mayo Clinic, Rochester, MN

BACKGROUND

- Current HR-pQCT protocols do not account for limb length despite tremendous variation in bone density and geometry along the radius and tibia.
- Consequently, limb length adds considerable variability that confounds the interpretation of biological effects and increases scatter in cross-sectional and normative data.

AIMS

- To measure the variability in bone quality parameters due to limb length
- Propose a protocol for scanning anatomically-standard regions based on measured limb-length.

LIMB MEASUREMENTS

- Limb length was measured by palpating superficial anatomic features, marking their location on the skin, and measuring the distance between using anthropometric tape
- Tibial length was measured between the tibial malleolous on the distal end and tibial plateau on the proximal end **(precision: RMSE = 0.7 mm, 1.9%)**
- Forearm length measured using the ulna: between the styloid process on the distal end and end of the elbow on the proximal end **(precision: RMSE = 0.3 mm, 1.1%)**

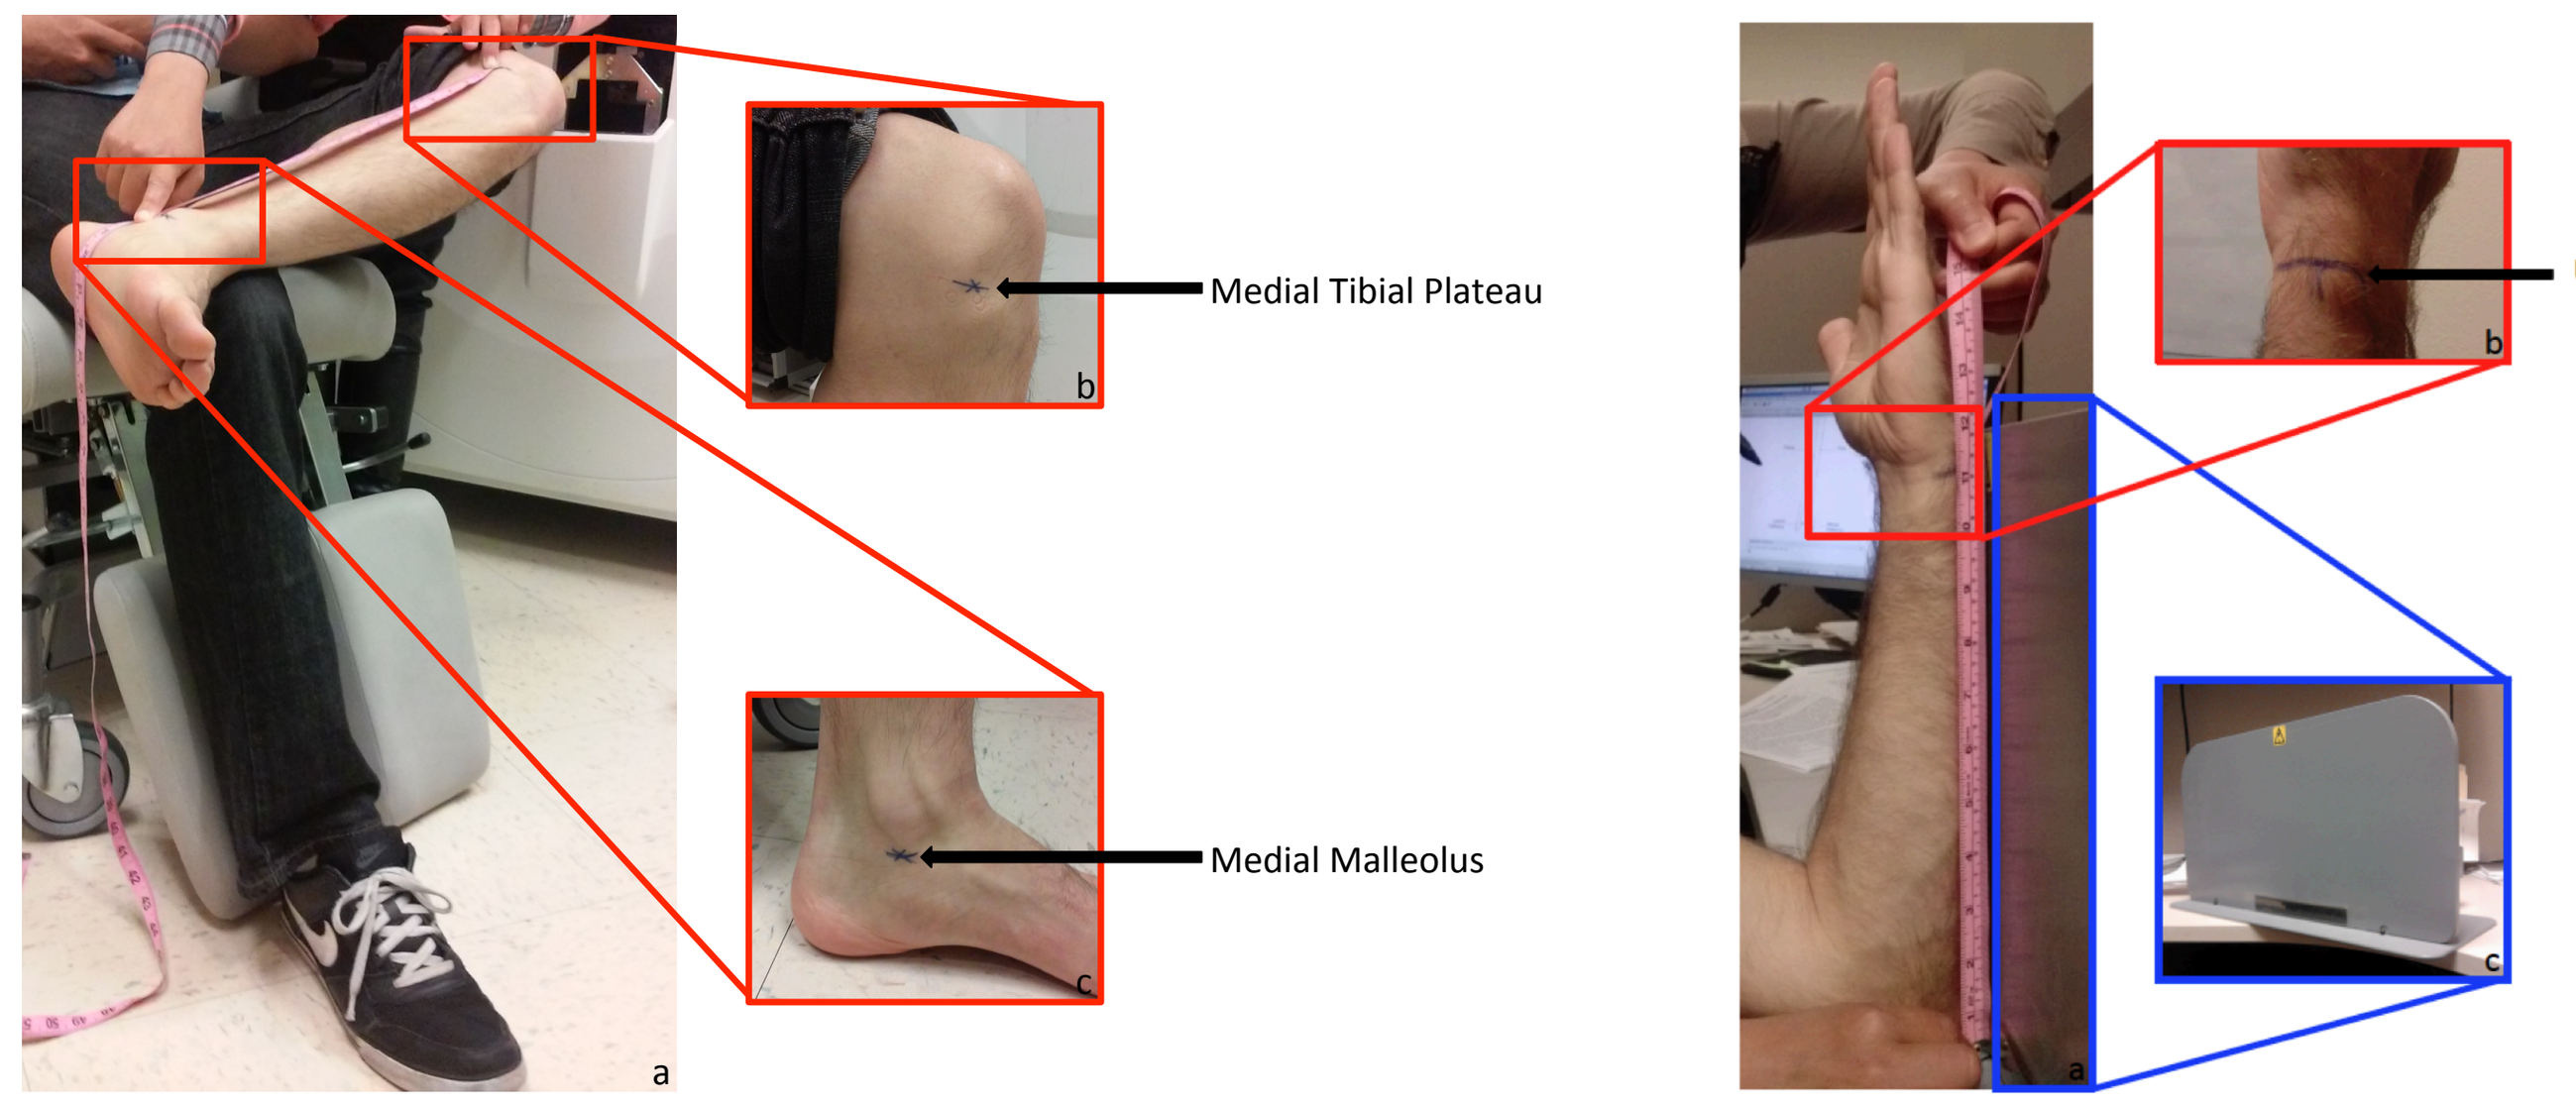
Medial Tibial Plateau
Medial Malleolus
Ulnar Styloid Process
a
b
c

Figure 1. Limb length measurement of the lower leg (left) and forearm (right)

IMAGING EXPERIMENTS

- Double stack (220-slice) HR-pQCT scans of the distal radius and tibia were acquired in 30 adults (17 men, 13 women)
- Bone measurements for 110-slice sub-volumes were extracted for volumes corresponding to a scan positioned at:
 I. The default fixed offset location
 II. The average anatomic position (% of total limb length) of the study population
 III. The estimated average anatomic position (% of total limb length) of normative populations reported in the literature
- Mean and SD of differences between fixed and anatomic positions calculated for BMD, Tb.BMD, Ct.Th, Tb.N

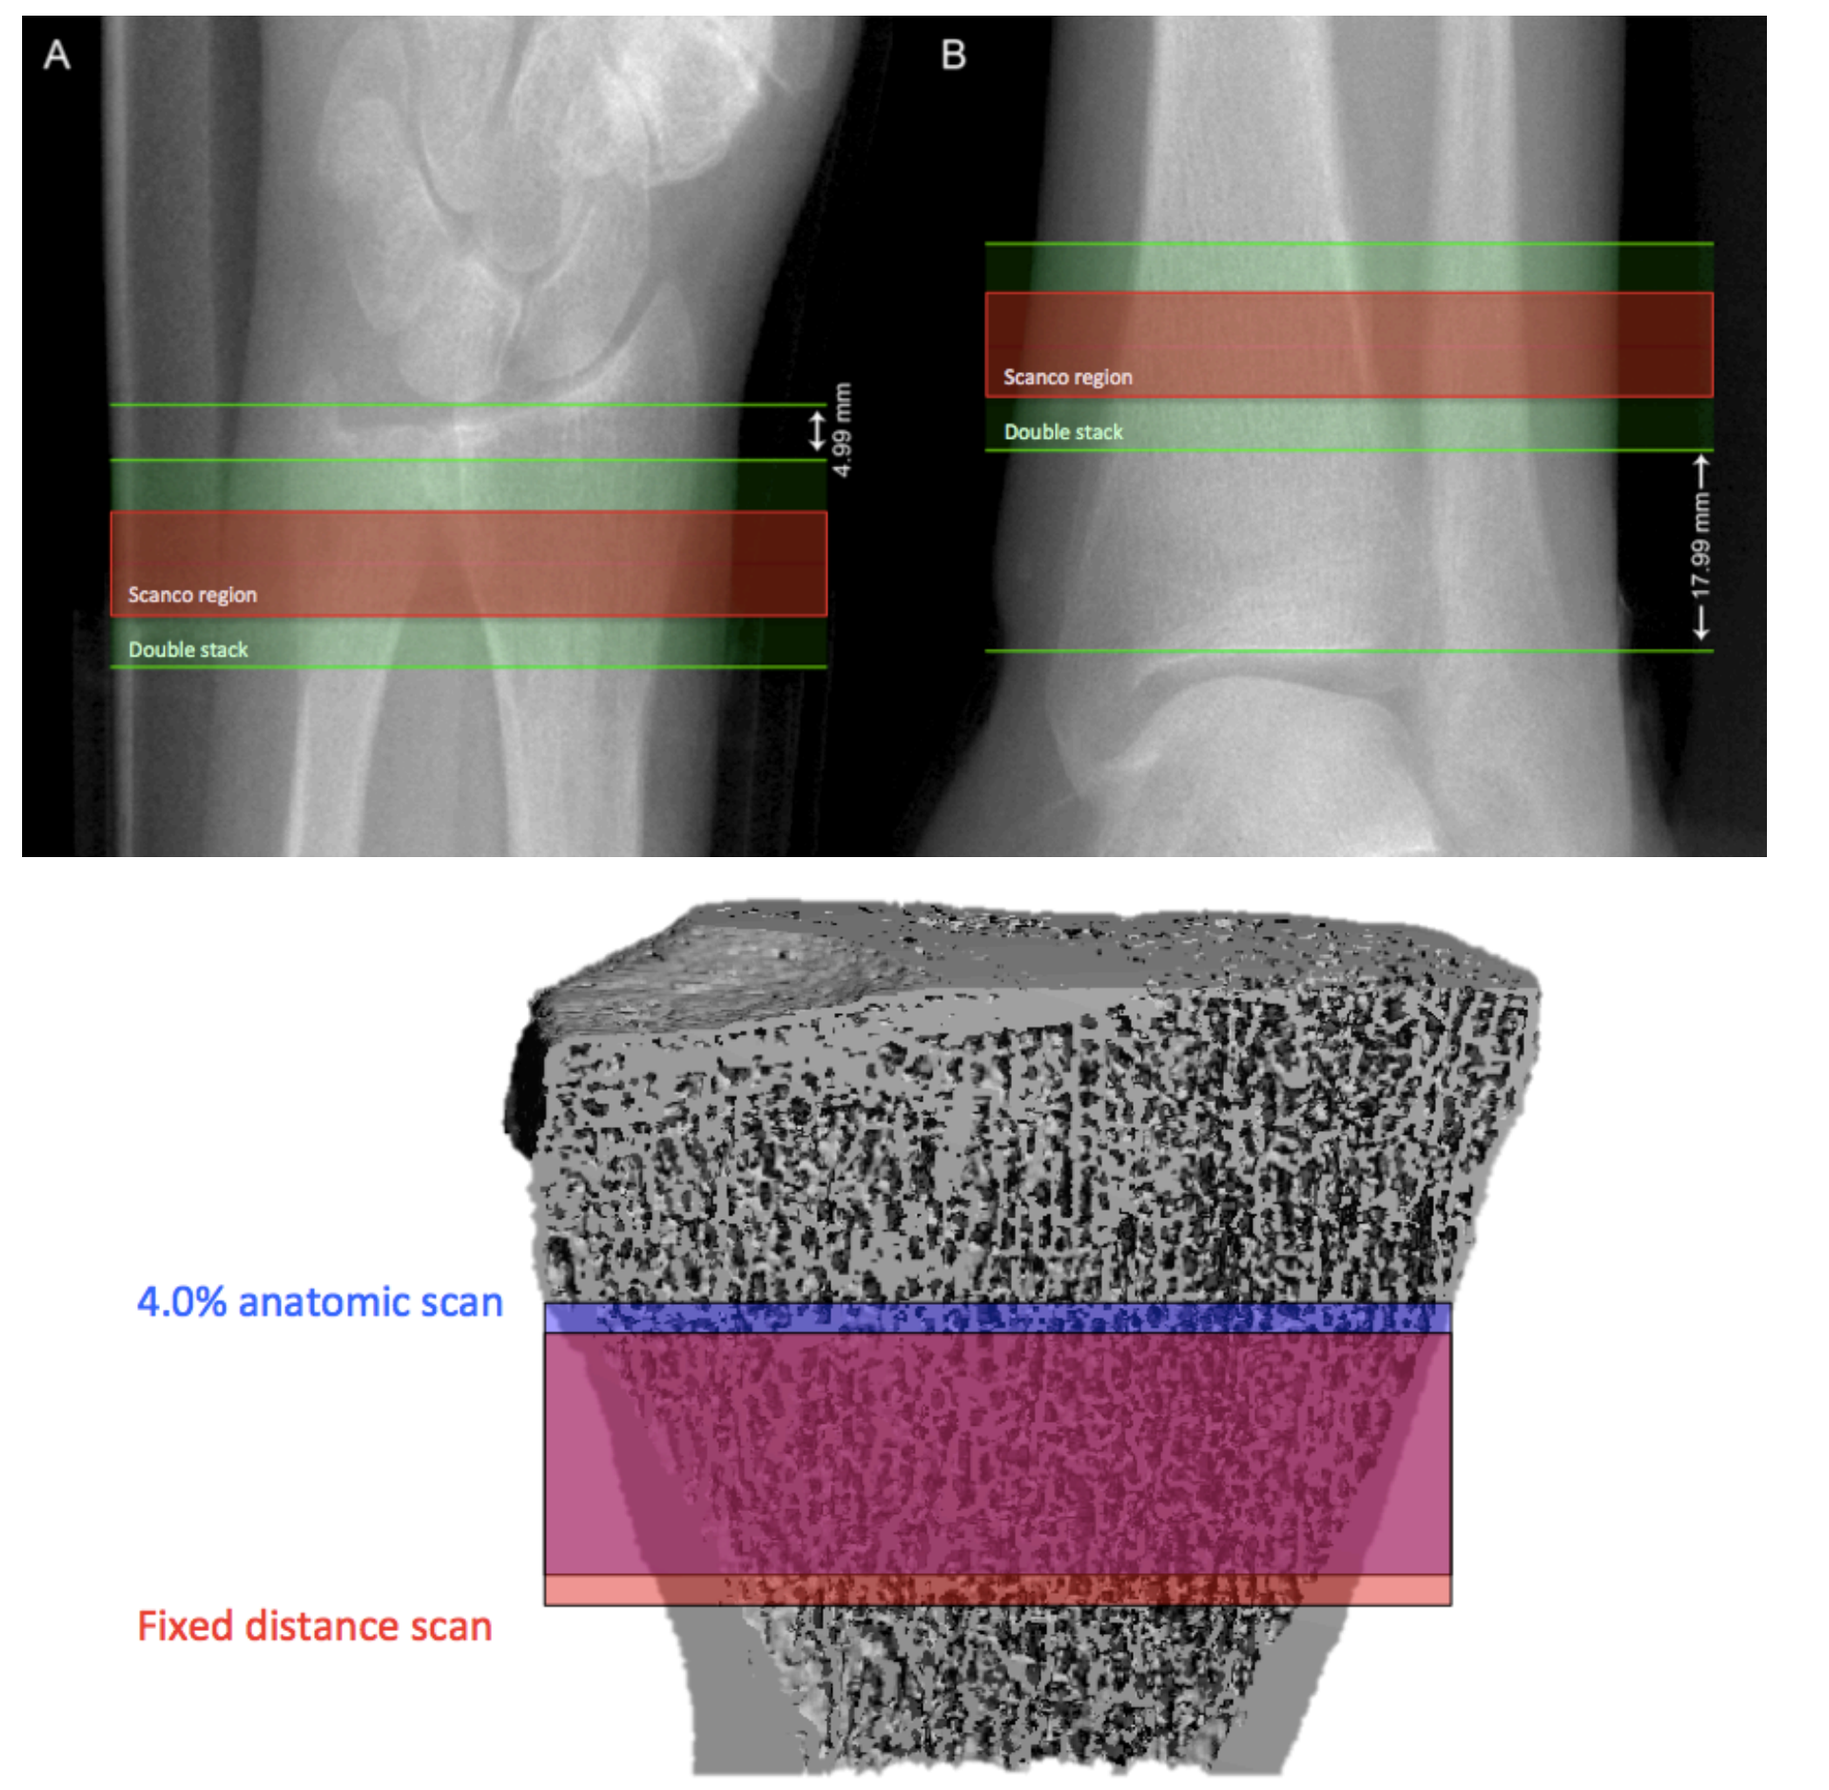
A
B
Scanco region
Double stack
4.99 mm
17.99 mm
4.0% anatomic scan
Fixed distance scan

Figure 2. Scout view of double-stack scan site (top) and illustration of sub-volume analysis (bottom)

RESULTS

- Average anatomic position in the study population
 - **Radius: 4.0±0.5% (± 1.1 mm)**
 - **Tibia: 7.2±0.6% (± 2.2 mm)**
- Backwards-Compatible anatomic position based on the literature survey:
 - **Radius: 4.1%**
 - **Tibia: 7.4%**

Table 1. Variability in HR-pQCT measures due to limb Length

	Relative position based on limb length measured in current study participants **[Radius: 4.0% \| Tibia: 7.2%]**		Relative position estimated from height of existing HR-pQCT population data in the literature **[Radius: 4.1% \| Tibia: 7.4%]**	
	Difference vs. Fixed: Population Averages	**Variance and Range of Differences**	**Difference vs. Fixed: Population Averages**	**Variance and Range of % Differences**
Radius				
BMD	-0.6%	6.7% [-9.8, +20.8]	1.2%	6.6% [-10.2, +19.5]
Ct.BMD	0.4%	5.6% [-5.7, +20.2]	0.0%	5.4% [-6.2, +19.1]
Ct.Th	-0.9%	18.8% [-18.6, +66.7]	2.1%	18.6% [-19.8, +66.7]
Tb.BMD	0.5%	2.4% [-5.3, +5.1]	-0.5%	2.5% [-5.2, +5.6]
Tb.N	-0.6%	5.2% [-15.0, +3.4]	0.4%	5.2% [-14.6, +3.5]
Tibia				
BMD	-0.1%	3.1% [-6.0, +6.6]	-1.2%	3.2% [-7.4, +5.4]
Ct.BMD	0.1%	2.3% [-3.9, +5.4]	-0.7%	2.3% [-5.1, +5.0]
Ct.Th	-0.3%	11.8% [-24.1, +27.5]	-3.4%	11.5% [-28.6, +22.2]
Tb.BMD	0.2%	4.0% [-11.6, +7.4]	1.3%	4.3% [-10.9, +9.8]
Tb.N	-0.4%	5.0% [-20.4, +8.3]	0.6%	4.6% [-16.1, +10.4]

Estimation of Backwards-Compatible Relative Scan Site

- Literature survey of height statistics from normative HR-pQCT data[1-8]
- Derived linear models for height to limb-length relationships based on the current study population:

Men (N=2032)
AvgHeight=172.8cm

Women (N=1767)
AvgHeight=161.3cm

AvgHeight = 167.1cm

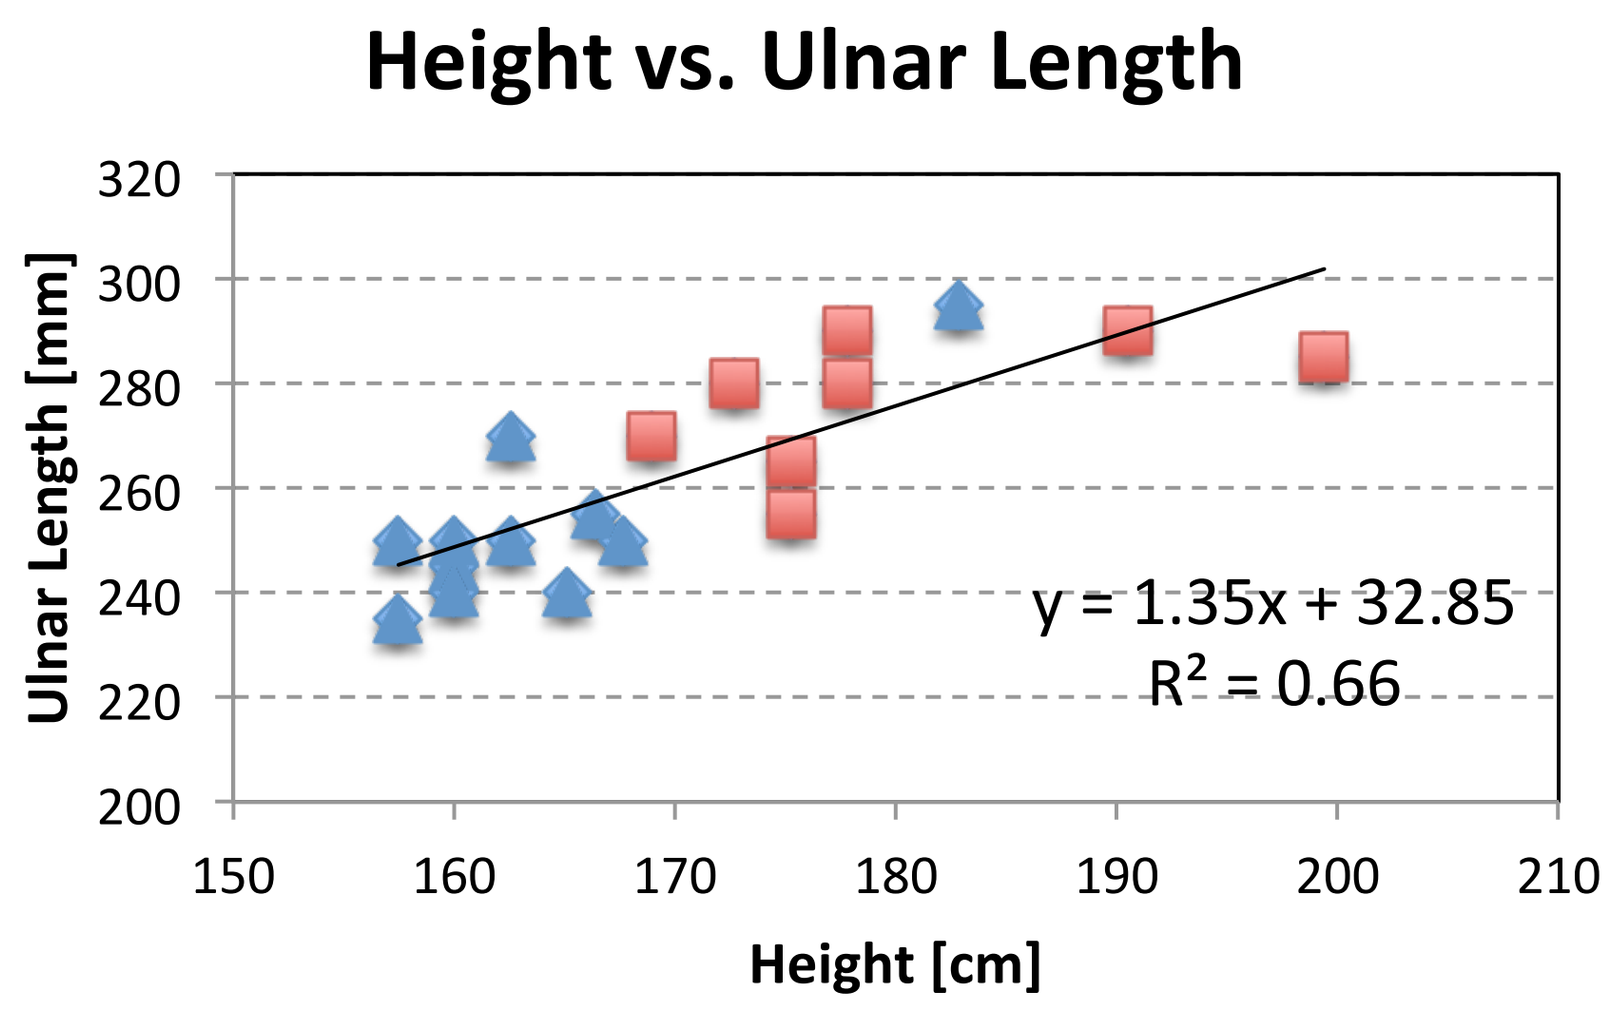
Height vs. Ulnar Length
Ulnar Length [mm]
Height [cm]
320
300
280
260
240
220
200
150
160
170
180
190
200
210
y = 1.35x + 32.85
R² = 0.66

Est. Avg. Ulnar Length = 25.8 cm

Est. Avg. Scan Position = 4.07%

[1]Dalzell, N., et al. (2009) [2]Burghardt, A. J., et al. (2010) [3]Nilsson, M., et al. (2010) [4]Macdonald, et al. (2011) [5]Szulc, P., et al. (2011) [6]Sornay-Rendu, E., et al. (2012) [7]Chevalley, T., et al. (2013) [8]Hansen, S., et al. (2013)

CONCLUSIONS

- The difference in bone measures from fixed vs. anatomically standard regions is the same or greater than errors introduced by patient motion
- This variability is strongest for cortical geometry and integral density measures, and less pronounced for trabecular bone measures
- Cross-sectional comparisons between populations with different height/limb-lengths are subject to significant biases when measured from scans made at fixed offsets
- For optimal backwards comparability with existing normative population data, standard anatomic positions of 4.1% for the radius, and 7.4% for the tibia should be used.

ACKNOWLEDGMENTS

The project is funded by **NIH/NIAMS R01 AR060700**

andrew.burghardt@ucsf.edu