Standardized Training For HR-pQCT Scan Positioning Reduces Inter-Operator Precision Errors

S. Bonaretti[1], N. Vilayphiou[2], C. M. Chan[3], A. Yu[1], M. Holets[4], K.Nishiyama[5], D. Liu[6], S. Boutroy[7], A. Ghasem-Zadeh[8], S. K. Boyd[9], R. Chapurlat[7], H. McKay[6], E. Shane[5], M. L. Bouxsein[10], T. F. Lang[1]. S. Khosla[4], P. M. Cawthon[11]. D. M. Black[12], S. Majumdar[2], E. S. Orwoll[13], A. J. Burghardt[1]

[1]MQIR, Department of Radiology & Biomedical Imaging, University of California San Francisco, CA, USA; [2]Scanco Medical AG, Brüttisellen, Switzerland; [3]University of California, Berkeley, CA, USA; [4]Mayo Clinic, Rochester, MN, USA; [5]Metabolic Bone Diseases Unit, Columbia University Medical Center, New York, NY, USA; [6]University of British Columbia, Vancouver, BC, Canada, [7]INSERM UMR 1033, Université de Lyon, France; [8]University of Melbourne, Melbourne, Australia; [9]University of Calgary, Calgary, Canada; [10]Beth Israel Deaconess Medical Center, Boston, MA, USA; [11]California Pacific Medical Center Research Institute, San Francisco, CA, USA; [12]University of California, San Francisco, CA, USA; [13]Oregon Health & Science University, Portland, OR, USA

BACKGROUND

- HR-pQCT is an *in-vivo* imaging technique used to assess bone quality, evaluate bone diseases and monitor drug therapies
- The operator acquires a projection of the limb, and s/he visually identifies an anatomical landmark that determines the region to be scanned
- Variability in landmark identification impacts bone measurements, especially in the radius, and affects data comparability in multicenter studies

AIM

- To compare intra- and inter-operator precision for experienced operators without training vs. new operators with training
- To develop a training and certification platform (webapp) for HR-pQCT operators

PRECISION EXPERIMENT

- We reproduced the acquisition interface of the HR-pQCT system (XtremeCT, Scanco Medical AG)
- We used scout-view images corresponding to double-stack (220 slices) HR-pQCT scans
- We virtually localized standard 110-slice volume based on each operator's positioning
- New operators underwent training with theoretical and practical demonstration, and simulated scan positioning exercises

OPERATOR PRECISION

- A total of 8 experienced and 6 new operators position reference lines at anatomical landmarks
- We measured positioning precision and impact on bone parameters for short-term intra-operator and inter-operator reproducibility

RADIUS	**Precision** ***SD_{RMS} [mm]***	**Impact on Bone Parameter Measurements (*CV_{RMS} [%]*)** BMD	Ct.BMD	Tb.BMD	Ct.Th	Tb.N
Short-term intra-op						
Experienced	0.24	1.39	0.93	0.40	3.17	0.47
New	0.28	1.50	1.26	0.72	3.46	0.69
Inter-op						
Experienced	0.68	3.69	2.56	1.46	8.40	2.12
New	0.34	2.09	1.50	0.87	4.90	1.32
Scan/rescan	-	0.89	0.76	0.72	2.10	4.23

TIBIA	**Precision** ***SD_{RMS} [mm]***	**Impact on Bone Parameter Measurements *CV_{RMS} [%]*** BMD	Ct.BMD	Tb.BMD	Ct.Th	Tb.N
Short-term intra-op						
Experienced	0.13	0.26	0.19	0.26	0.94	0.31
New	0.11	0.31	0.30	0.28	0.52	0.31
Inter-op						
Experienced	0.30	0.61	0.42	0.65	1.97	0.85
New	0.16	0.30	0.21	0.32	1.02	0.41
Scan/rescan	-	0.43	1.38	0.70	1.25	5.64

DISCUSSION

- Inter-operator variability can be significantly reduced with standardized training
- To make our training platform available to the community, we developed a webapp

WEBAPP

http://webapps.radiology.ucsf.edu/msk/

- Landing page with information and documentation

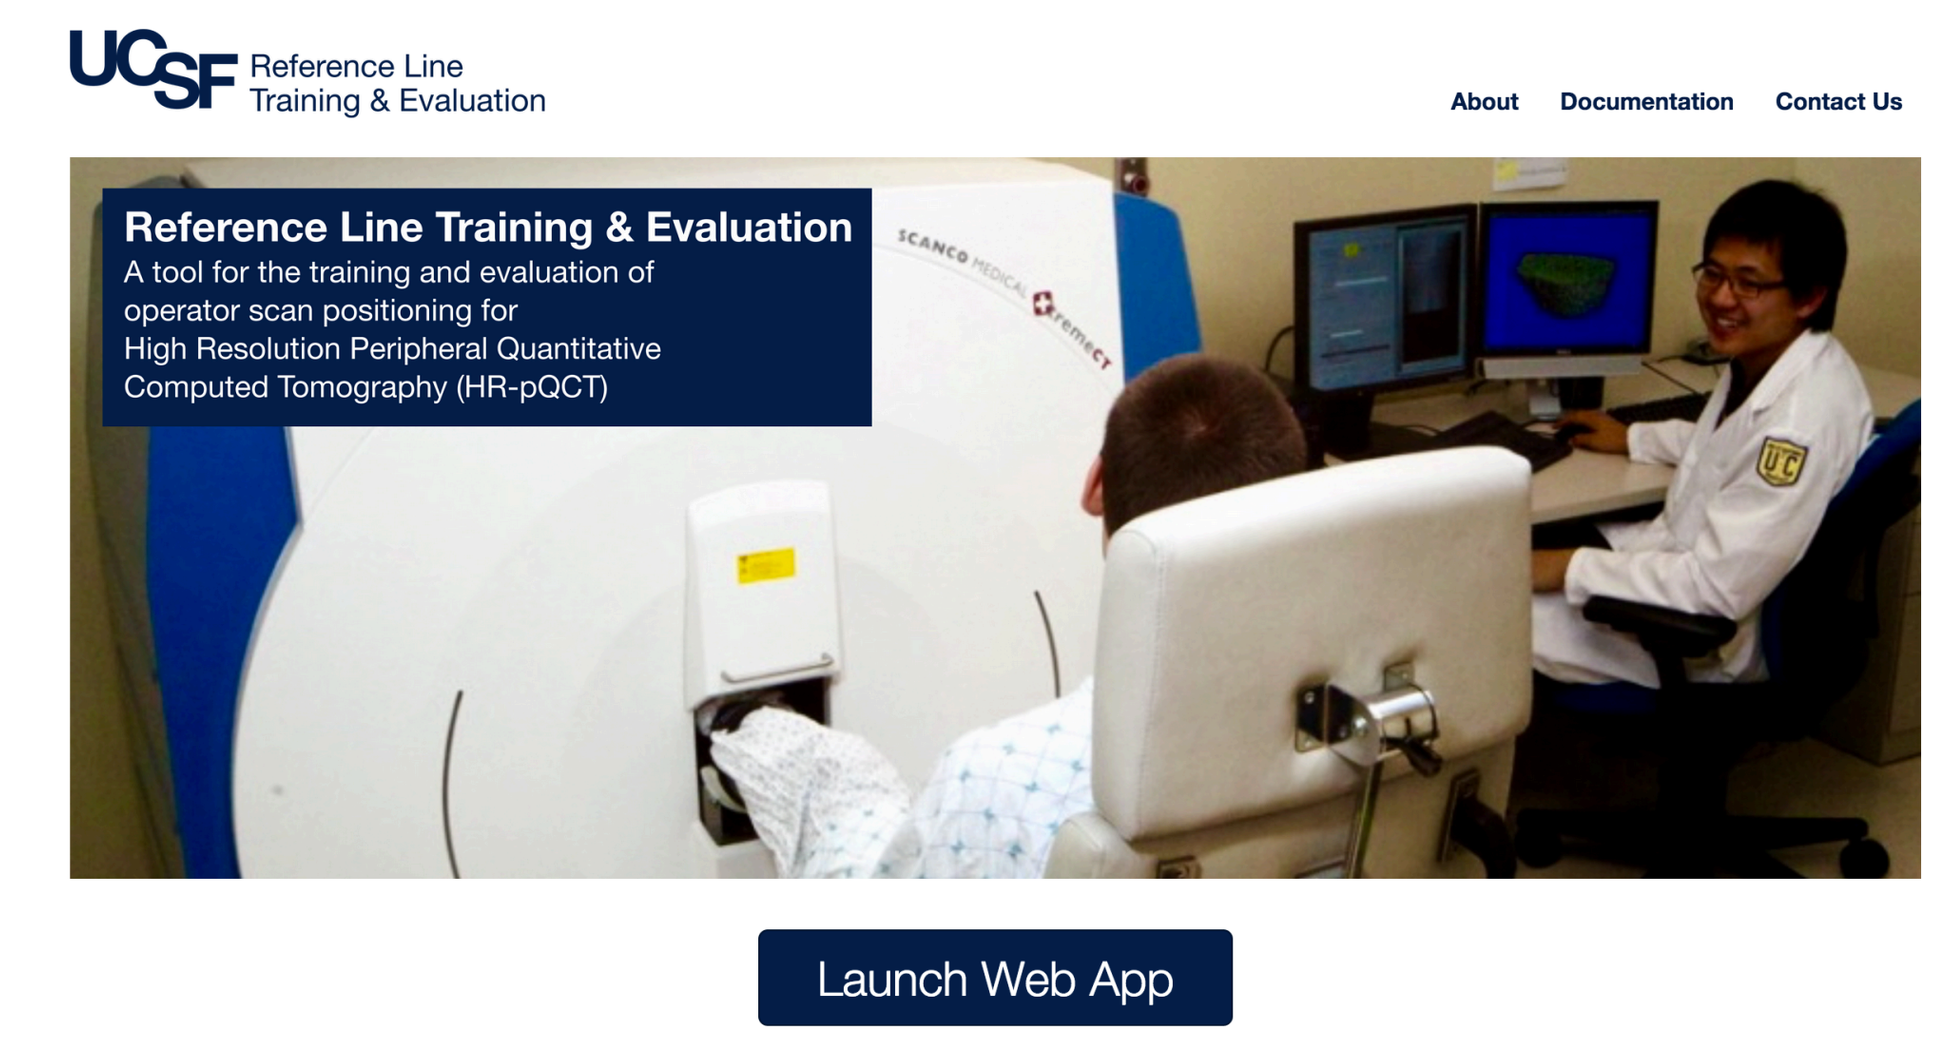
Reference Line
Training & Evaluation
About
Documentation
Contact Us
Reference Line Training & Evaluation
A tool for the training and evaluation of operator scan positioning for High Resolution Peripheral Quantitative Computed Tomography (HR-pQCT)
Launch Web App

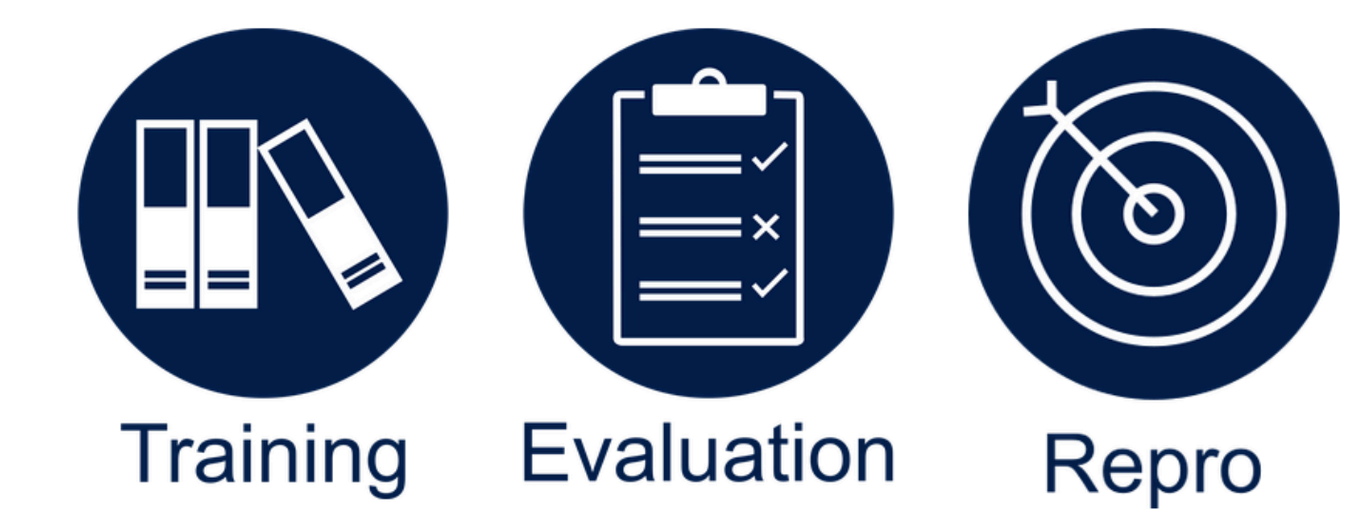
Training
Evaluation
Repro

- Modules simulate scanner acquisition software
- For radius and tibia scans
- Training Module for practice on a selectable variety of typical cases
 - Images grouped by anatomical features
 - Feedback window presented at the completion of each set
- Evaluation Module for certification
 - For certification, must pass three randomly presented sets
- Reproducibility Module for operator precision test
 - Visual feedback of position variability and with a summary precision metric (RMSE) reported.

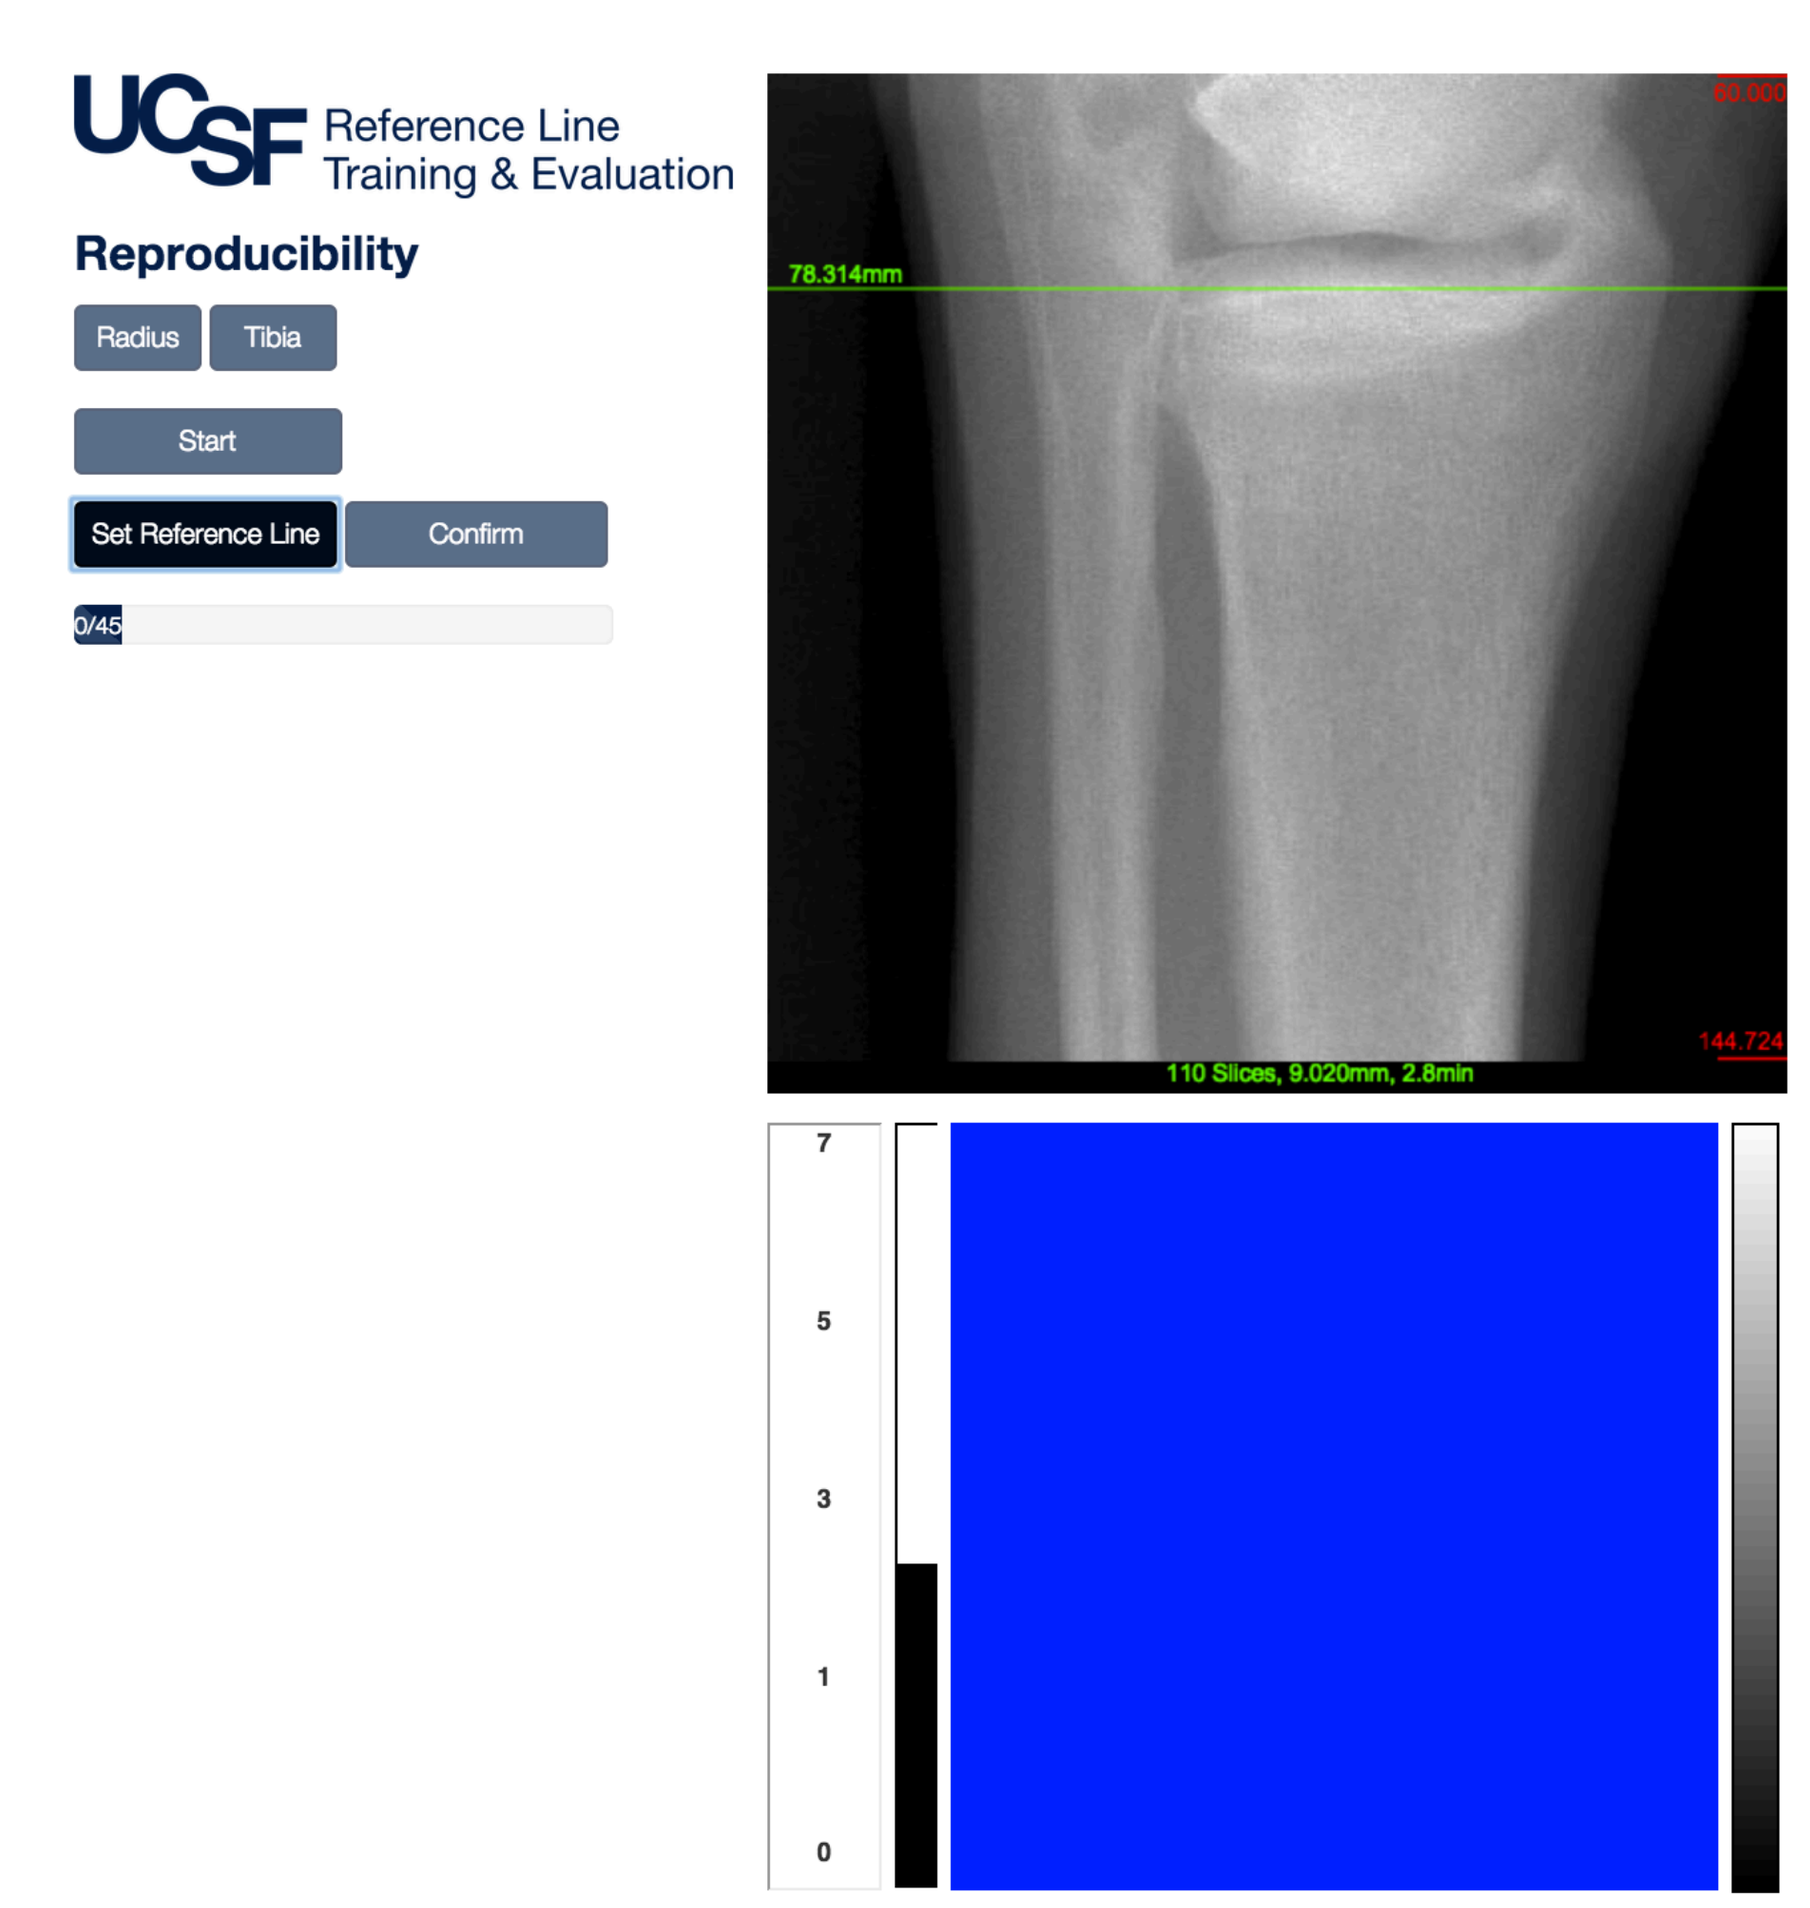
Reference Line
Training & Evaluation
Reproducibility
Radius
Tibia
Start
Set Reference Line
Confirm
0/45
78.314mm
110 Slices, 9.020mm, 2.8min

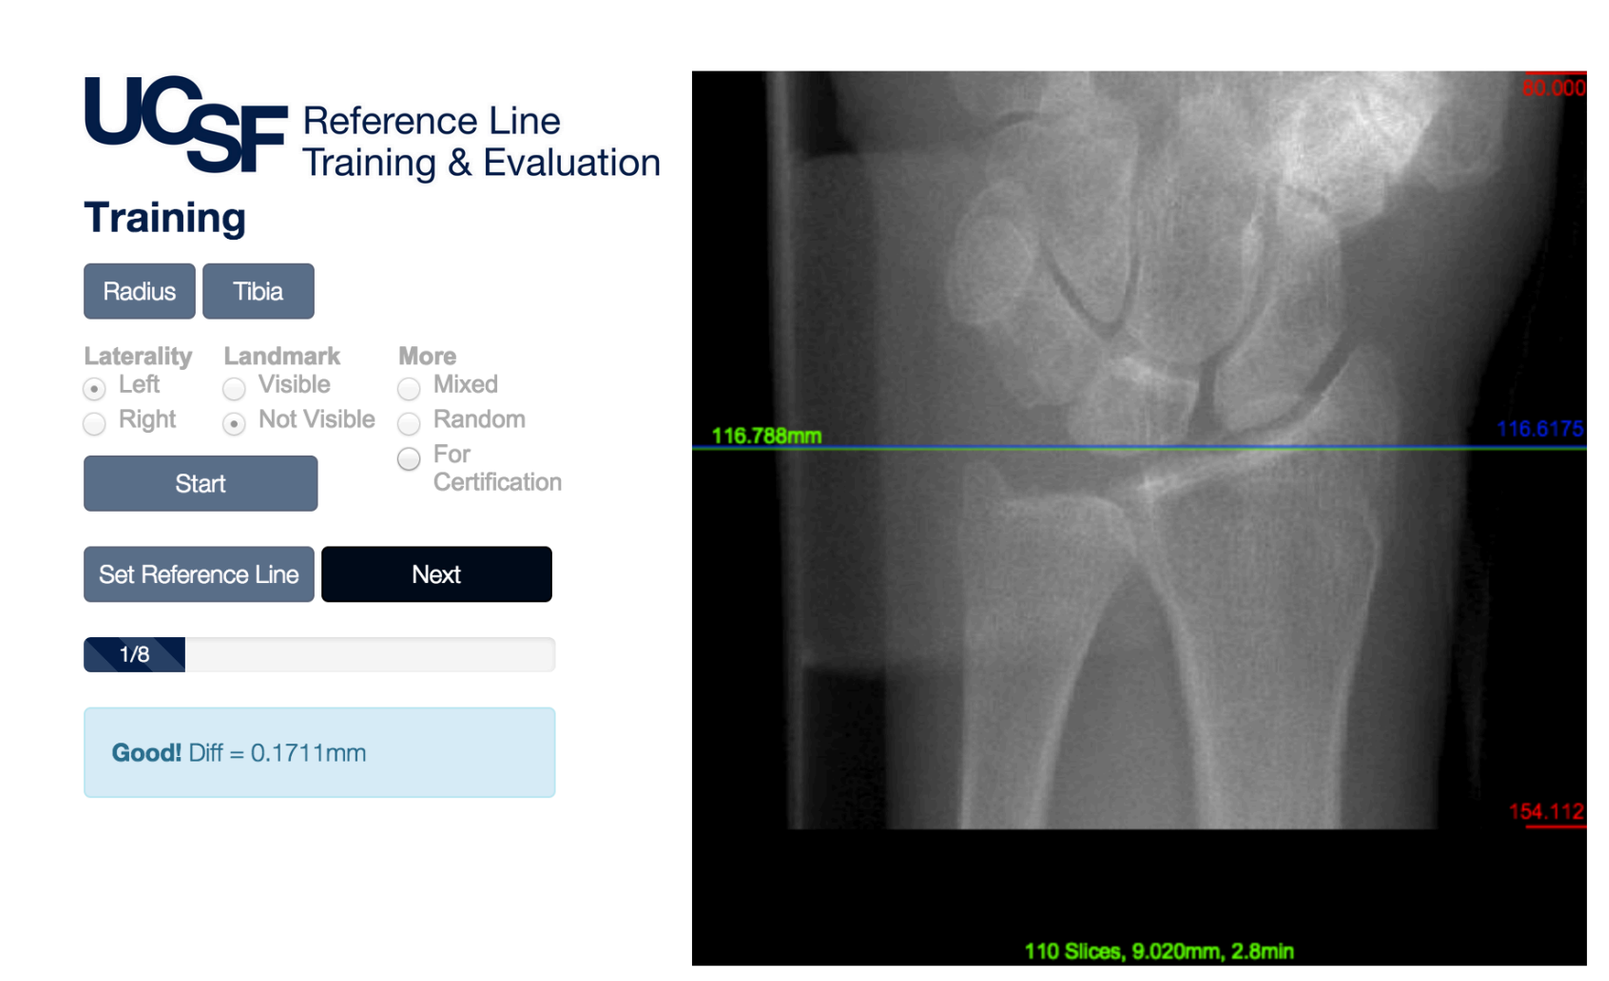
Reference Line
Training & Evaluation
Training
Radius
Tibia
Laterality
Left
Right
Landmark
Visible
Not Visible
More
Mixed
Random
For Certification
Start
Set Reference Line
Next
1/8
Good! Diff = 0.1711mm
116.788mm
110 Slices, 9.020mm, 2.8min

Top: Immediate feedback after confirming line placement in Training module

Left: Reproducibility Module in the Set Reference Line stage

The project is funded by NIH/NIAMS 5R01AR060700 and by NIH/NIA-NIAMS-NCATS U01AG027810, U01AG042124, U01AG042139, U01AG042140, U01AG042143, U01AG042145, U01AG042168, U01AR066160, UL1TR000128